Refining The Measurement Of Motor And Non-Motor Symptoms in Parkinson's Disease Patients With Fluctuating Symptoms

Philip O. Buck[1], Michelle K. White[2], Jane Castelli-Haley[1], Regina Rendas-Baum[2]

[1]Teva Neuroscience, [2] i3 QualityMetric

BACKGROUND

- Parkinson's disease (PD) research and clinical management has traditionally focused on motor symptoms, such as tremors and postural stability, but study findings [1,2,3], strongly suggest that non-motor symptoms, such as anxiety and cognitive impairment, are key indicators of patients' health-related quality of life (HRQOL).
- Therapy-induced symptom fluctuations (*wearing off* and *on-off* fluctuations) are common after a few years of treatment. Hence, a major aim in the management of PD patients is to reduce the amount of daily symptom fluctuations and the time spent in the *off* state, i.e. when symptoms are present.
- Measurement of *off* time and symptom fluctuations are commonly captured by *on/off* diaries kept by patients. The Scales for Outcomes in Patients with Parkinson's Disease Diary Card (SCOPA-DC) [4] is a *on/off* diary designed to measure both the severity and the stability of problems with motor function in PD patients with fluctuating symptoms.
- The SCOPA-DC was validated in a sample of PD patients in the Netherlands but has not been validated in a U.S. population. In addition, the SCOPA-DC does not assess non-motor symptoms.

OBJECTIVES

- Establish the content validity of the SCOPA-DC in a U.S. population.
- Determine the feasibility of adding items/domains that measure non-motor functions.

METHODS

LITERATURE REVIEW

OBJECTIVES

- Identify the most dominant non-motor symptoms experienced by PD patients with fluctuating symptoms.
- Review the patient reported outcome (PRO) instruments currently recommended for measurement of non-motor symptoms.

SEARCH

- Medline on PubMed was searched for relevant papers using the text words "rating scale", "nonmotor", "non-motor", "daily diary", "on/off", "on-off", "quality of life", "SCOPA" in combination with ''Parkinson's,'' or ''Parkinson".

FOCUS GROUPS

OBJECTIVES

- Determine whether the SCOPA-DC was well comprehended and relevant in a U.S. population.
- Obtain patient input on the non-motor symptoms that are most relevant to PD patients with fluctuating symptoms.
- Test the content validity of the revised SCOPA-DC.

RECRUITMENT

A total of 33 patients were recruited to participate in 3 focus groups (N=8 per focus group) and one cognitive debrief (N=9). Focus group participants were recruited according to the following PD-related criteria: ≥ 30 years of age; presence of *off* time symptoms; at least 2 of the following 3 symptoms: 1) slowed ability to start and continue movements (bradykinesia), 2) resting tremors or shakiness, and 3) rigidity (inability to complete a movement or stiffness); currently taking at least one of the following PD medications: Levadopa (Larodopa®), Carbidopa (Lodosyn®), Levadopa/Carbidopa (Sinemet®, Sinemet CR®), Levodopa/Carbidopa/Entacapone (Stalevo®); never had brain surgery to treat PD; fluent in English.

RESULTS: LITERATURE REVIEW

- Findings from the literature review supported the addition of non-motor symptoms to the SCOPA-DC, which were shown to have a greater impact on HRQOL than motor symptoms, and were associated with reports of experiencing fluctuating symptoms.
- Although a few PD-specific PRO instruments [5,6] included health domains beyond motor function, these instruments were not designed for multiple daily self-reported assessments.
- The following non-motor symptoms were identified as the most relevant to PD patients with fluctuations: 1) feelings of anxiety; 2) mood swings; 3) loss of interest; 4) fatigue; 5) autonomic/ gastrointestinal symptoms (excessive sweating, salivation, and incontinence).

RESULTS: FOCUS GROUPS

PARTICIPANT CHARACTERISTICS

- Participants were mostly Caucasian (82%), male (67%) and retired (70%). Three quarters experienced *off* time between 1% and 25% of the time. Nearly half of the participants had been diagnosed with PD for more than 5 years (Table 1).

Original SCOPA-DC

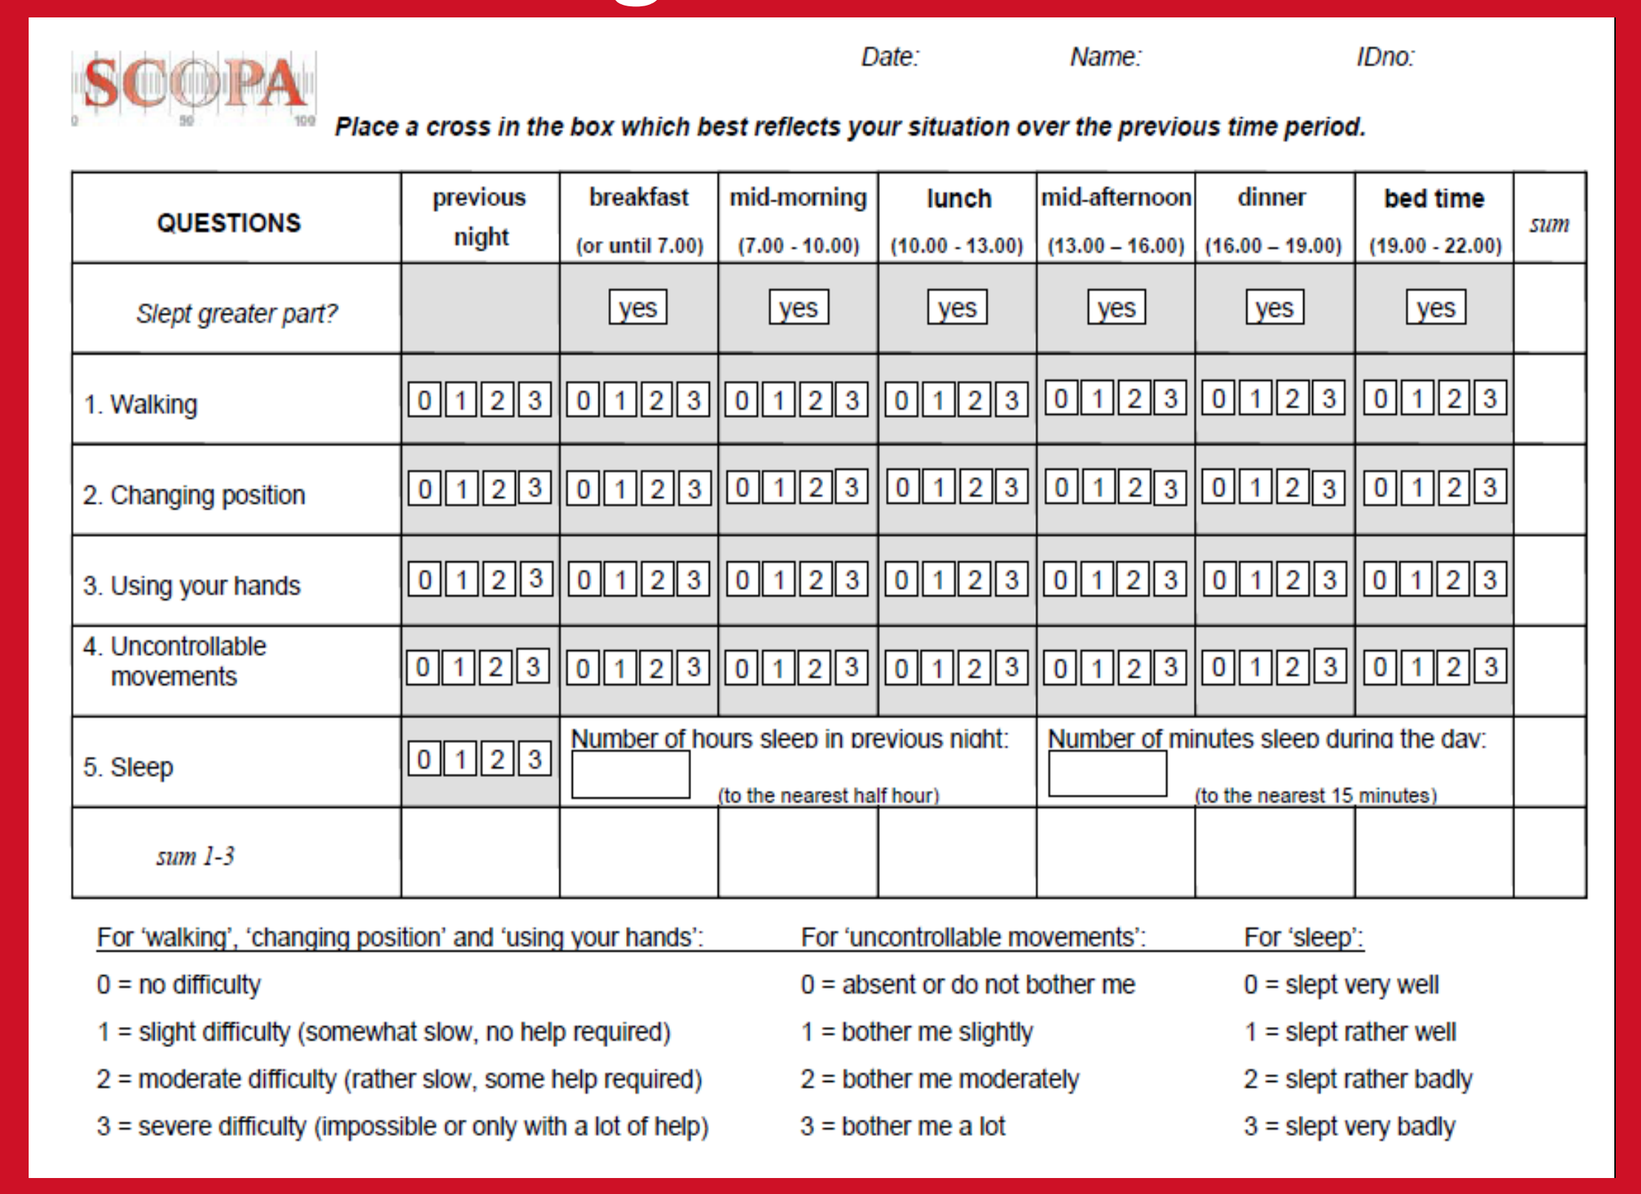

SCOPA Date: Name: IDno:

Place a cross in the box which best reflects your situation over the previous time period.

QUESTIONS	previous night	breakfast (or until 7.00)	mid-morning (7.00 - 10.00)	lunch (10.00 - 13.00)	mid-afternoon (13.00 – 16.00)	dinner (16.00 – 19.00)	bed time (19.00 - 22.00)	sum
Slept greater part?		yes	yes	yes	yes	yes	yes	
1. Walking	0 1 2 3	0 1 2 3	0 1 2 3	0 1 2 3	0 1 2 3	0 1 2 3	0 1 2 3	
2. Changing position	0 1 2 3	0 1 2 3	0 1 2 3	0 1 2 3	0 1 2 3	0 1 2 3	0 1 2 3	
3. Using your hands	0 1 2 3	0 1 2 3	0 1 2 3	0 1 2 3	0 1 2 3	0 1 2 3	0 1 2 3	
4. Uncontrollable movements	0 1 2 3	0 1 2 3	0 1 2 3	0 1 2 3	0 1 2 3	0 1 2 3	0 1 2 3	
5. Sleep	0 1 2 3	Number of hours sleep in previous night: (to the nearest half hour)			Number of minutes sleep during the day: (to the nearest 15 minutes)			
sum 1-3								

For 'walking', 'changing position' and 'using your hands': 0 = no difficulty; 1 = slight difficulty (somewhat slow, no help required); 2 = moderate difficulty (rather slow, some help required); 3 = severe difficulty (impossible or only with a lot of help)

For 'uncontrollable movements': 0 = absent or do not bother me; 1 = bother me slightly; 2 = bother me moderately; 3 = bother me a lot

For 'sleep': 0 = slept very well; 1 = slept rather well; 2 = slept rather badly; 3 = slept very badly

Revised SCOPA-DC

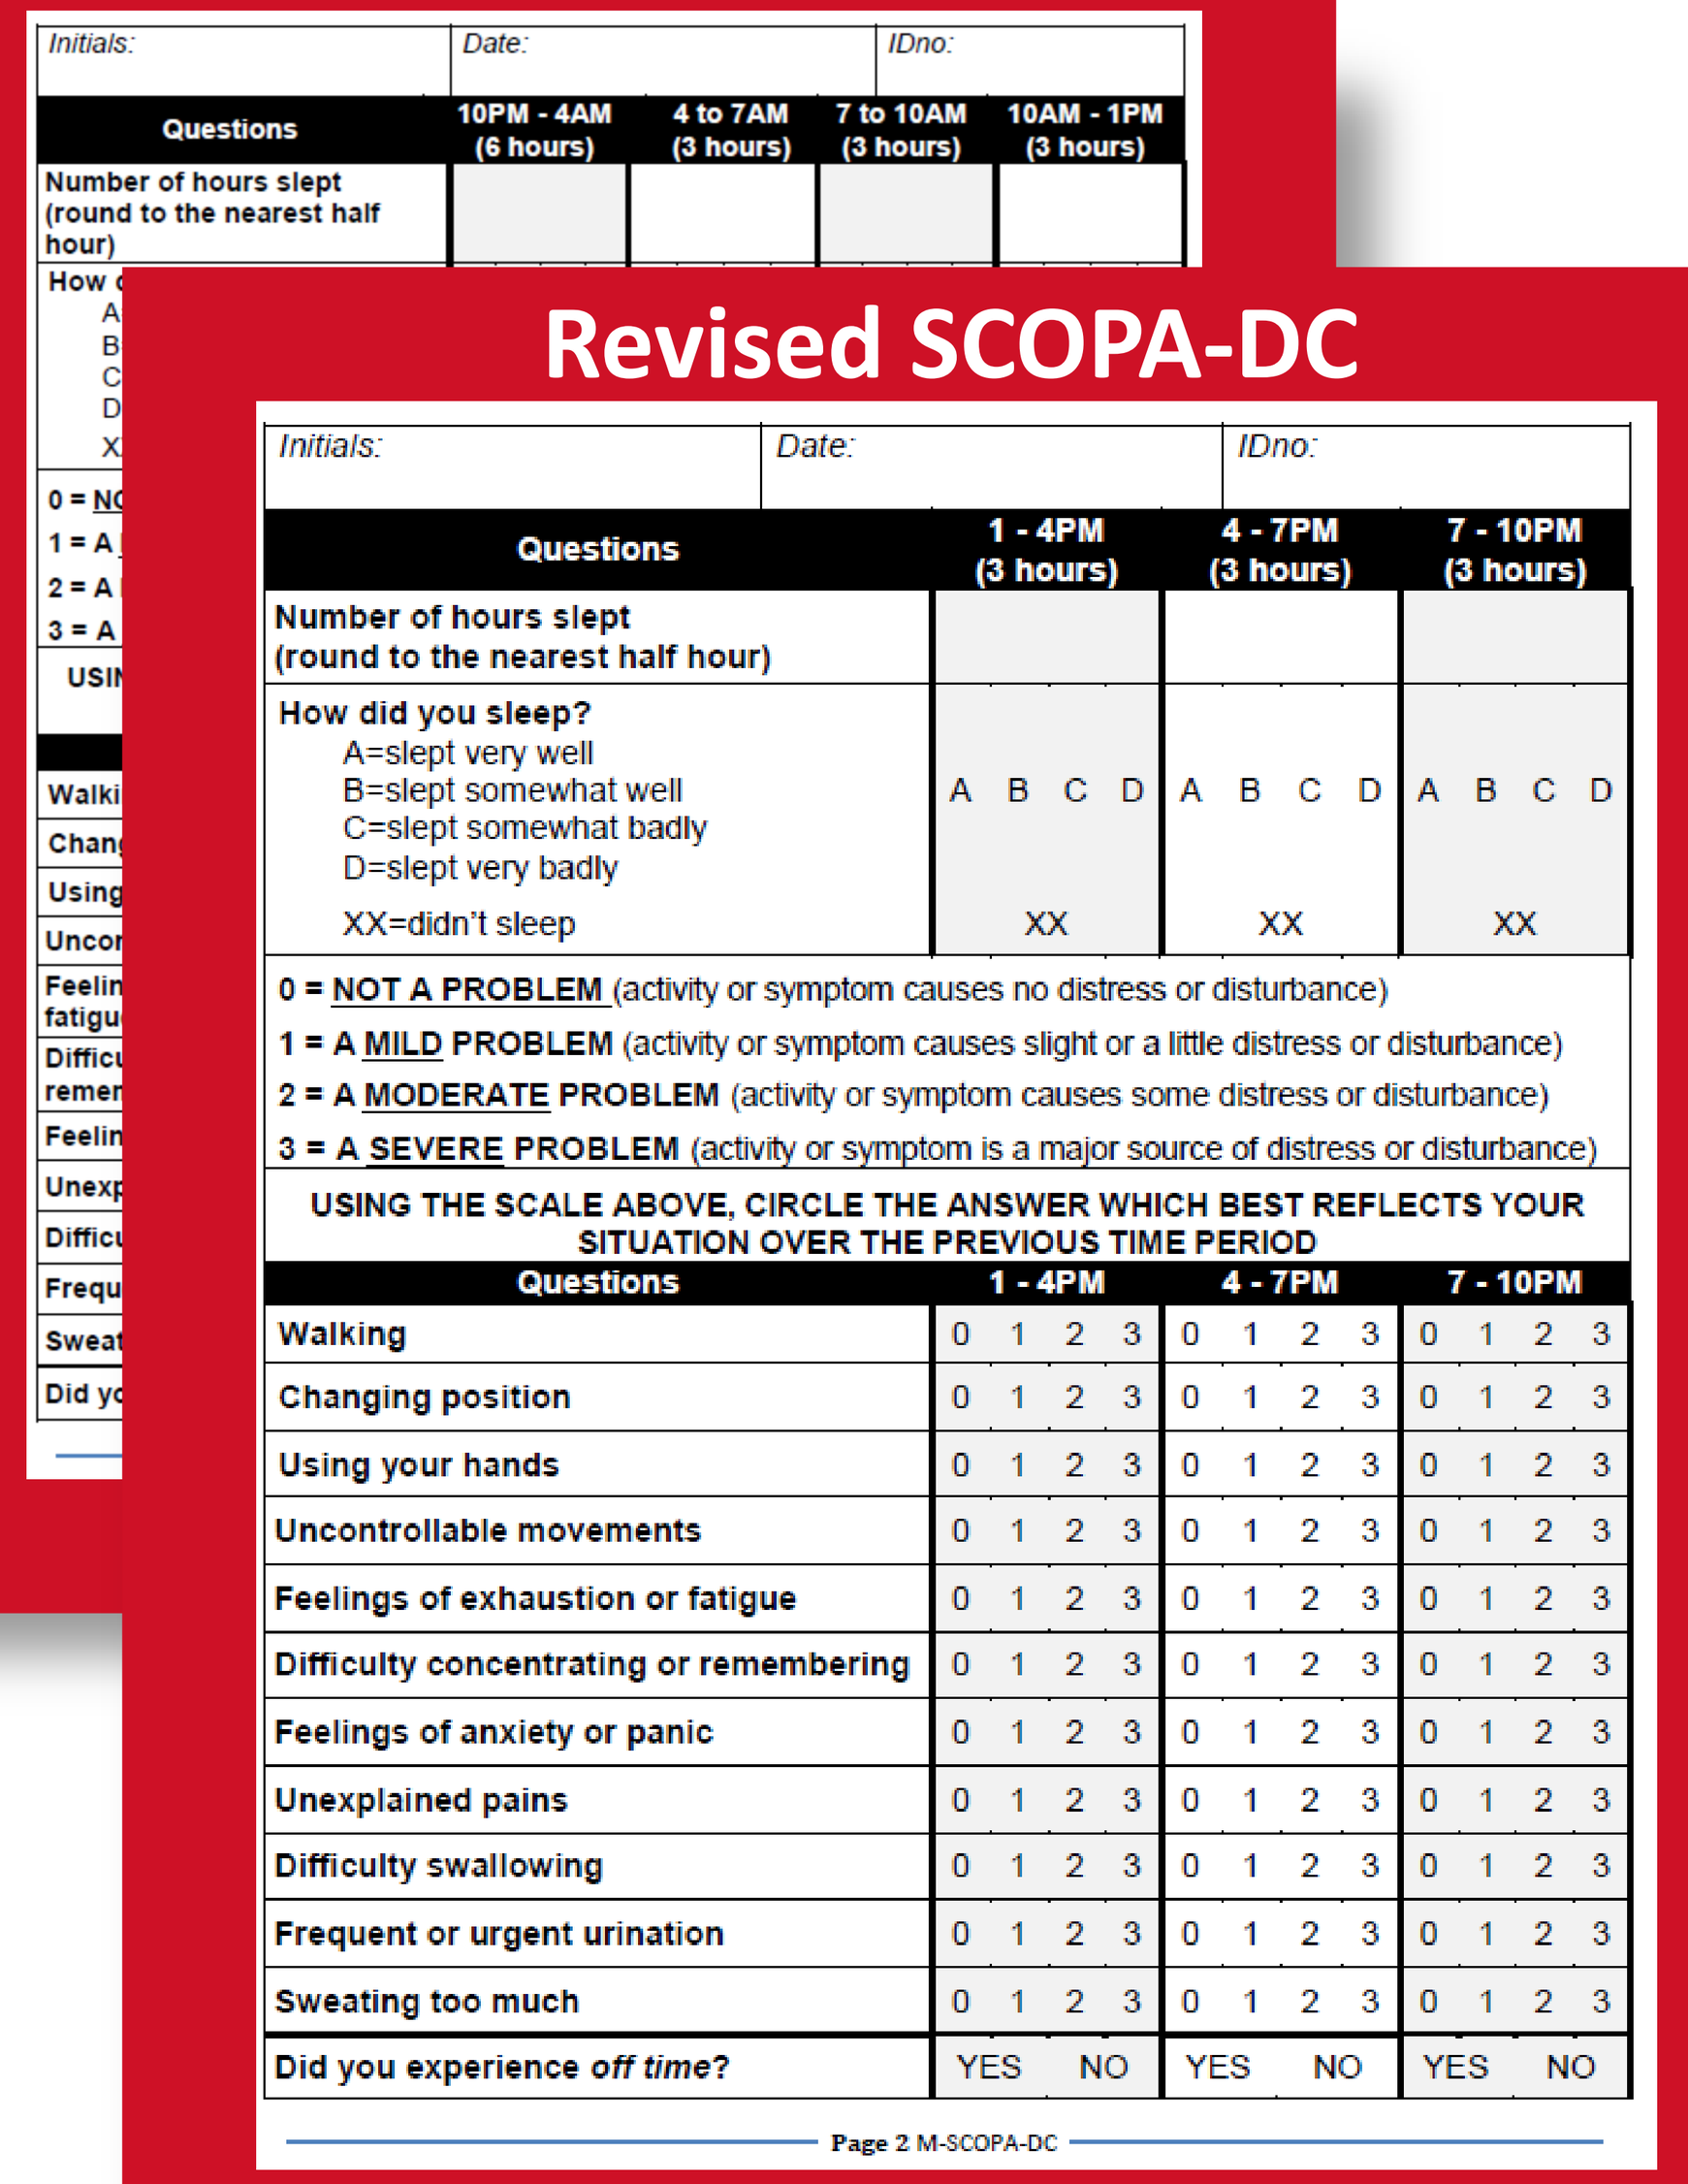

Initials: Date: IDno:

Questions	10PM - 4AM (6 hours)	4 to 7AM (3 hours)	7 to 10AM (3 hours)	10AM - 1PM (3 hours)
Number of hours slept (round to the nearest half hour)				

Initials: Date: IDno:

Questions	1 - 4PM (3 hours)	4 - 7PM (3 hours)	7 - 10PM (3 hours)
Number of hours slept (round to the nearest half hour)			
How did you sleep? A=slept very well B=slept somewhat well C=slept somewhat badly D=slept very badly XX=didn't sleep	A B C D XX	A B C D XX	A B C D XX

0 = NOT A PROBLEM (activity or symptom causes no distress or disturbance)
1 = A MILD PROBLEM (activity or symptom causes slight or a little distress or disturbance)
2 = A MODERATE PROBLEM (activity or symptom causes some distress or disturbance)
3 = A SEVERE PROBLEM (activity or symptom is a major source of distress or disturbance)

USING THE SCALE ABOVE, CIRCLE THE ANSWER WHICH BEST REFLECTS YOUR SITUATION OVER THE PREVIOUS TIME PERIOD

Questions	1 - 4PM	4 - 7PM	7 - 10PM
Walking	0 1 2 3	0 1 2 3	0 1 2 3
Changing position	0 1 2 3	0 1 2 3	0 1 2 3
Using your hands	0 1 2 3	0 1 2 3	0 1 2 3
Uncontrollable movements	0 1 2 3	0 1 2 3	0 1 2 3
Feelings of exhaustion or fatigue	0 1 2 3	0 1 2 3	0 1 2 3
Difficulty concentrating or remembering	0 1 2 3	0 1 2 3	0 1 2 3
Feelings of anxiety or panic	0 1 2 3	0 1 2 3	0 1 2 3
Unexplained pains	0 1 2 3	0 1 2 3	0 1 2 3
Difficulty swallowing	0 1 2 3	0 1 2 3	0 1 2 3
Frequent or urgent urination	0 1 2 3	0 1 2 3	0 1 2 3
Sweating too much	0 1 2 3	0 1 2 3	0 1 2 3
Did you experience *off time*?	YES NO	YES NO	YES NO

Page 2 M-SCOPA-DC

Table 1. Characteristics of focus group participants (N=33)

Age, mean (min/max)	69 (49/80)
Male, n (%)	22 (67%)
Ethnicity, n (%)	
White, non-Hispanic	27 (82%)
Hispanic	5 (15%)
Asian	1 (3%)
Education, n (%)	
High school graduate or GED	2 (6%)
Associate's / Technical degree	15 (45%)
College graduate /Graduate degree	16 (49%)
Employment Status, n (%)	
Not employed / on disability	7 (21%)
Employed part-time / full-time	3 (9%)
Retired	23 (70%)
Living Situation, n (%)	
Living with spouse or partner	29 (88%)
Living with friends or family	3 (9%)
Living alone	1 (3%)
Time since PD diagnosis, n (%)	
Less than 2 years	6 (18%)
Between 2 and 5 years	12 (36%)
More than 5 years	15 (46%)
Percentage of *off* time, n (%)	
1-25%	24 (73%)
26-50%	6 (18%)
51-75%	2 (6%)
76-100%	1 (3%)

References

1. Cheon, S. M., Ha, M. S., Park, M. J., & Kim, J. W. (2008). Nonmotor symptoms of Parkinson's disease: prevalence and awareness of patients and families. Parkinsonism Relat. Disord., 14, 286-290.
2. Simuni, T. & Sethi, K. (2008). Nonmotor manifestations of Parkinson's disease. Ann.Neurol., 64 Suppl 2, S65-S80.
3. Snyder, C. H. & Adler, C. H. (2007). The patient with Parkinson's disease: part I-treating the motor symptoms; part II-treating the nonmotor symptoms. J.Am.Acad.Nurse Pract., 19, 179-197.
4. Marinus, J., Visser, M., Stiggelbout, A. M., Rabey, J. M., Bonuccelli, U., Kraus, P. H. et al. (2002). Activity-based diary for Parkinson's disease. Clin.Neuropharmacol., 25, 43-50.
5. Stacy M.A., Murphy J.M., Greeley D.R., Stewart R.M., Murck H., Meng X (2008). The sensitivity and specificity of the 9-item Wearing-off Questionnaire. Parkinsonism Relat .Disord ., 14, 205-212.
6. Chaudhuri, K. R., Martinez-Martin, P., Brown, R. G., Sethi, K., Stocchi, F., Odin, P. et al. (2007). The metric properties of a novel non-motor symptoms scale for Parkinson's disease: Results from an international pilot study. Mov. Disord., 22, 1901-1911.

RESULTS: FOCUS GROUPS

VALIDITY OF CONTENT AND FORMAT OF ORIGINAL SCOPA-DC

- Findings from the 3 focus groups demonstrated support for the content of the original SCOPA-DC, generally finding it to be clear and intuitive, despite difficulties with some item definitions, time frames, and response labels.

DOMAIN	PATIENT'S TESTIMONY
Sleep Time: Participants had difficulty understanding the sleep question in the original SCOPA-DC (*Slept greater part?*) and indicated they were uncertain why they did not have a response option for every day segment including the previous night.	*Slept the greater part it says yes here […] what do you do?... just X in the column? … So you don't mark the first one at all?* [when asked what would be better:]*… Just put when you go to sleep, how long and when you wake up.*
Day Segment Evaluation: Participants struggled to find congruence between the time frames and day segment labels. Further, many indicated that they relied on the day segment labels rather than the time frames to account for their PD experiences throughout the day, while others used the time frames only.	*I think at the top it would be better to just have the hours, not breakfast, mid-morning, lunch.*
Response Option Evaluation: Participants indicated that they did not use the specific response items (labels) as directed. Rather than checking which question set was related to a particular response option scale, they simply used a generalized 4-point scale.	*If the zero meant either the difference between no difficulty and absent, I say take that column away no matter what column you look at, I'm going to put it at zero.*
Item Evaluation: All, but one, of the original SCOPA-DC items were found to be clear and intuitive. Some patients indicated that their responses would have included tremors when considering the uncontrollable movements item.	*It's tremors, it's a shaking of your muscles inside you can feel you can't see it because my hands aren't shaking like this.*

DOMAIN ELICITATION

- Participants spoke about the emotional effects of PD and identified non-motor symptoms that complicated their ability to complete activities of daily life or to engage in work or social situations.

DOMAIN	PATIENT'S TESTIMONY
Fatigue: Fatigue often had an unexpected onset followed by inability to do anything but rest.	*I don't think the doctors understand how tired we can feel, it's an overwhelming tiredness not just little tiredness. Like you really can't do anything, you can't get out of the chair.*
Concentration/Memory: Participants expressed frustration with their inability to recall simple facts or events.	*Reading a column in the Chicago Tribune, I read the first three lines and I go off into the sunset and stop reading for no reason at all, my body tells me I can't digest anymore , and it completely stops.*
Anxiety: The physical challenges of being in public were often the precursor to feelings of anxiety.	*[…] I don't want to create an incident. I don't want to have the ambulance come and pick me up and take me to the hospital. I get that guilt.*
Pain: Most participants explained that they would be awakened by pains during the night or when resting.	*My right leg, first thing in the morning it will wake me up. When I'm sleeping.*
Difficulty Swallowing: Participants felt that their mouth and throat muscles didn't function well enough to swallow food.	*I have got to be real careful because the food will go down the wrong way and then I'll be choking for the next 15 minutes.*
Frequent Urination: Participants spontaneously informed focus groups' staff about the need for frequent and often uncontrollable restroom breaks.	*[…] I'm not able to drink anything before I go to bed because I have to get up 4 or 5 times.*
Sweating: tasks that were once relatively easy, such as walking while shopping, could cause enough exertion to break into a sweat, which caused embarrassment.	*Well first off I go shopping and I'm beginning to sweat and I can feel it in my legs, I am making an effort walking. So, I can get all sweaty.*

CONCLUSION

Our findings indicated that the inclusion of non-motor symptoms in a PD diary was greatly valued by patients. Meaningful time frames and clarity of response choices were also important factors to patients. A reliable and validated patient-reported daily diary may improve the ability to describe PD progression by accurately measuring both motor and non-motor symptoms. Additional quantitative research is needed to evaluate the psychometric properties of the revised SCOPA-DC.

Presented at the ISPOR, 15th Annual International Meeting, May 17-20, Atlanta, GA. This study was supported by Teva Neuroscience. For additional information, please contact Philip Buck, (816) 508-8446 or Philip.Buck@tevaneuro.com