Confirmatory Factor Analysis and Differential Item Functioning Analysis of the Migraine-Specific Quality of Life Questionnaire Version 2.1 in Adults with Chronic Migraine

Regina Rendas-Baum, MS[1], Gregory A Maglinte, PhD[2], Michael De Rosa, MA[1], Min Yang, MD, PhD[1], Sepideh F Varon, PhD[2]

[1]i3 QualityMetric, Lincoln, RI; [2]Allergan, Inc., Irvine CA;

BACKGROUND

- Studies have shown that migraine can be a progressive disorder. Individuals with episodic migraine (EM; < 15 headache days per month [HDPM]) may progress to chronic migraine (CM; ≥ 15 HDPM) (1).
- A higher frequency of headache attacks is associated with significantly greater levels of disability and reduced health-related quality of life (HRQOL) (2).
- The Migraine-Specific Quality of Life Questionnaire Version 2.1 (MSQ) is a 14-item health-related quality of life instrument that measures the functional impact of migraine across three domains: Role Function-Restrictive (RR), Role Function-Preventive (RP) and Emotional Function (EF).
- Reliability and validity of the MSQ have been primarily studied in EM populations (3-5). Given the emerging evidence regarding the multiplicity of factors that distinguish CM from EM patients, studies assessing the reliability and validity of the MSQ specifically in CM patients are needed.

OBJECTIVES

To evaluate the factor structure and cross-cultural comparability of the MSQ in a sample of CM patients.

METHODS

STUDY DESIGN

- A web-based, cross-sectional, observational study of subjects who had previously reported suffering from headaches and/or migraines was conducted between February and April of 2009 in 9 countries: Australia, Canada, France, Germany, Italy, Spain, Taiwan, United Kingdom, and United States.
- Study eligibility criteria: 1) be 18 years of age or older; 2) have an active email address; 3) report having experienced headaches during the last 3 months not associated with a cold, the flu, a head injury or a hangover; 4) provide conformed consent by "opting-in" via a web link in the e-mail invitation; 5) have migraine symptoms (or probable migraine symptoms in the US only).
- Respondents were considered to have migraine if they reported all of the following: 1) at least one headache day in the last 3 months not associated with a cold, the flu, a head injury, or a hangover; 2) at least one type of headache (i.e., headaches that may differ in how they begin or the location of the pain); 3) at least one day of any type of headache in the past 3 months; 4) indicated experiencing moderate or severe pain "less than half the time" or "half the time or more"; 5) indicated experiencing nausea "less than half the time" or "half the time or more", or photophobia and phonophobia "less than half the time" or "half the time or more".
- Respondents were classified as having CM per ICHD-2 criteria with ≥15 headache days/month (n=499).

FACTOR STRUCTURE

- Confirmatory factor analysis (CFA) of the 3-factor model was conducted using the robust maximum likelihood estimator (MLR). Goodness-of-fit was assessed by the comparative fit index (CFI), Tucker-Lewis Index (TFI), and root mean square error of approximation (RSMEA).
- Three CFA models were evaluated:
 - 3-factor CFA, which mimics the currently proposed MSQ measurement model;
 - 1-factor model;
 - 3-factor model, in which MSQ item 12 is loaded onto the RR latent factor instead of EF.
- The 1-factor CFA was conducted as correlations between the latent factors were relatively high (.80's-.90's) indicating the potential for one overall, unidimensional factor.
- The 3-factor model where item 12 of the MSQ loaded on the RR latent factor was conducted as previous item-scale correlations showed that item 12 had a significantly higher correlation with the RR total score than with the EF total score.
- To examine item properties and their relationship with the hypothesized factor, item-scale correlations corrected for overlap, and item-removed Cronbach's alpha were evaluated.

DIFFERENTIAL ITEM FUNCTIONING (DIF)

- DIF was tested using ordinal logistic regression of MSQ item scores on group membership and trait level explanatory variables.
- Groups were based on country (Taiwan was excluded because of small sample size [N=8]) and language (English [Australia, Canada, Great Britain, United States] /non-English [Germany, Spain, France, Italy, Taiwan]).
- A significant (p<0.05) interaction term between group membership and trait level was indicative of non-uniform DIF. Uniform DIF was deemed to be present if the change in the trait level coefficient resulting from removal of the group membership term from the model was >10% (6).

RESULTS

SAMPLE CHARACTERISTICS (TABLE 1)

- Study recruitment is summarized in Figure 1.
- Study participants were mostly female (85.6%), White/Caucasian (89.4%), with a mean (±SD) age of 41.7 (±12.1). Nearly 80% of the total sample had been told by a healthcare provider that they suffered from migraines and the vast majority (92.4%) reported experiencing severe headache pain.
- Seven of the 9 countries were similarly represented with approximately 11% of the total sample each, while respondents from the United States formed approximately 20% of the total sample. Eight (1.6%) of the respondents were from Taiwan.
- Mean MSQ scores were highest for the RP scale (61.4; SD=26.1), followed by the EF scale (48.3; SD=28.1), and lowest for the RR scale (44.4; SD=22.1). No floor or ceiling effects were observed (Figure 2).

FACTOR STRUCTURE (TABLE 2)

- The 3-factor model demonstrated good fit (CFI = 0.97; TFI=0.96; RMSEA=0.07) among CM sufferers. Factor loadings ranged between 0.72 and 0.94, and had similar values across the three factors.
- An overall 1-factor CFA model, conducted to test unidimensionality, showed the worst model fit of the three models evaluated (CFI = 0.89; TFI=0.87; RMSEA=0.11).
- The model fit (CFI = 0.96; TFI=0.96; RMSEA=0.07) of the alternative 3-factor CFA where item 12 (*fed up or frustrated*) was moved to the RR factor was nearly identical to the proposed 3-factor model. However, the factor loading of item 12 on the RR latent factor was lower (0.67) than the factor loading on the EM factor (0.72).
- Item-scale correlations (Table 3) were highest for the hypothesized factor, with the exception of item 12, which correlated more strongly with the RR total score (0.67) than with the EF score (0.63). The impact of removing this item from the EF factor was however negligible, as Cronbach's alpha stayed essentially unchanged (Table 3).

DIFFERENTIAL ITEM FUNCTIONING (TABLES 4 AND 5)

- Most MSQ items showed absence of DIF. Non-uniform country DIF was identified in items 5 (*inability to concentrate*; p=0.028) and 12 (*fed up or frustrated*; p=0.037). Item 12 also presented non-uniform DIF related to language (p=0.010).

FIGURE 1. STUDY DESIGN SCHEMA

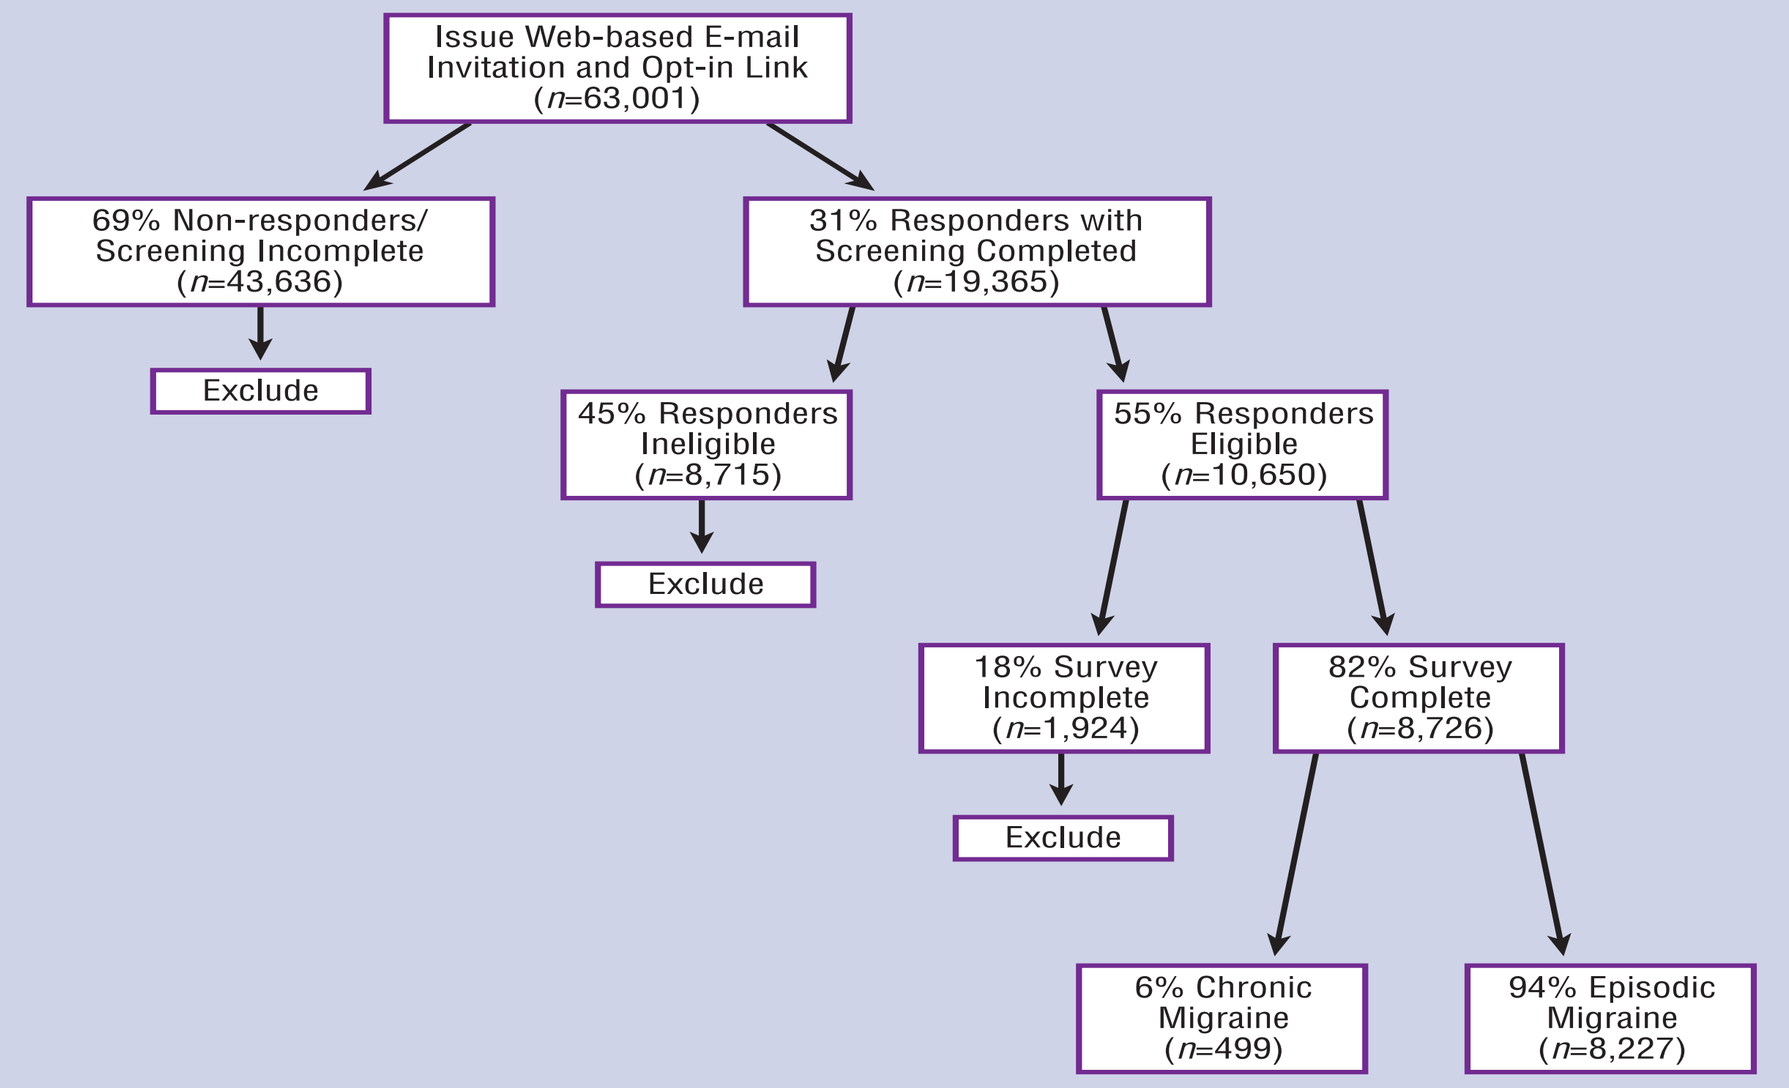
Issue Web-based E-mail Invitation and Opt-in Link (n=63,001)
69% Non-responders/ Screening Incomplete (n=43,636)
Exclude
31% Responders with Screening Completed (n=19,365)
45% Responders Ineligible (n=8,715)
Exclude
55% Responders Eligible (n=10,650)
18% Survey Incomplete (n=1,924)
Exclude
82% Survey Complete (n=8,726)
6% Chronic Migraine (n=499)
94% Episodic Migraine (n=8,227)

TABLE 1. SAMPLE CHARACTERISTICS (N=499)

Age, mean (SD)	41.7 (12.1)
Female, n (%)	427 (85.6%)
Race/ethnicity, n (%)	
White/Caucasian	446 (89.4%)
Asian	9 (1.8%)
Hispanic or Latino/Latin American	25 (5.0%)
Other	9 (1.8%)
Black	5 (1.0%)
Prefer not to answer	5 (1.0%)
Education, n (%)	
Less than a high school diploma	70 (14.0%)
High-school graduate	123 (24.6%)
Some college, no degree/ Associate degree	152 (30.5%)
Bachelor's degree	63 (12.6%)
Master's degree	28 (5.6%)
Doctoral degree	8 (1.6%)
Professional degree	33 (6.6%)
Other	16 (3.2%)
Prefer not to answer	6 (1.2%)
Healthcare provider told you that you suffer from migraines?, n (% yes)	394 (79.0%)
Headache pain severity, n (%)	
Mild	2 (0.4%)
Moderate	36 (7.2%)
Severe	461 (92.4%)
Comorbidities, n (%)	
Pain	208 (41.7%)
Psychiatric disorders	231 (46.3%)
Vascular disease events	41 (8.2%)
Vascular disease risk factors	208 (41.7%)
Other conditions	249 (49.9%)

FIGURE 2. MSQ SCALES DISTRIBUTION (N=499)

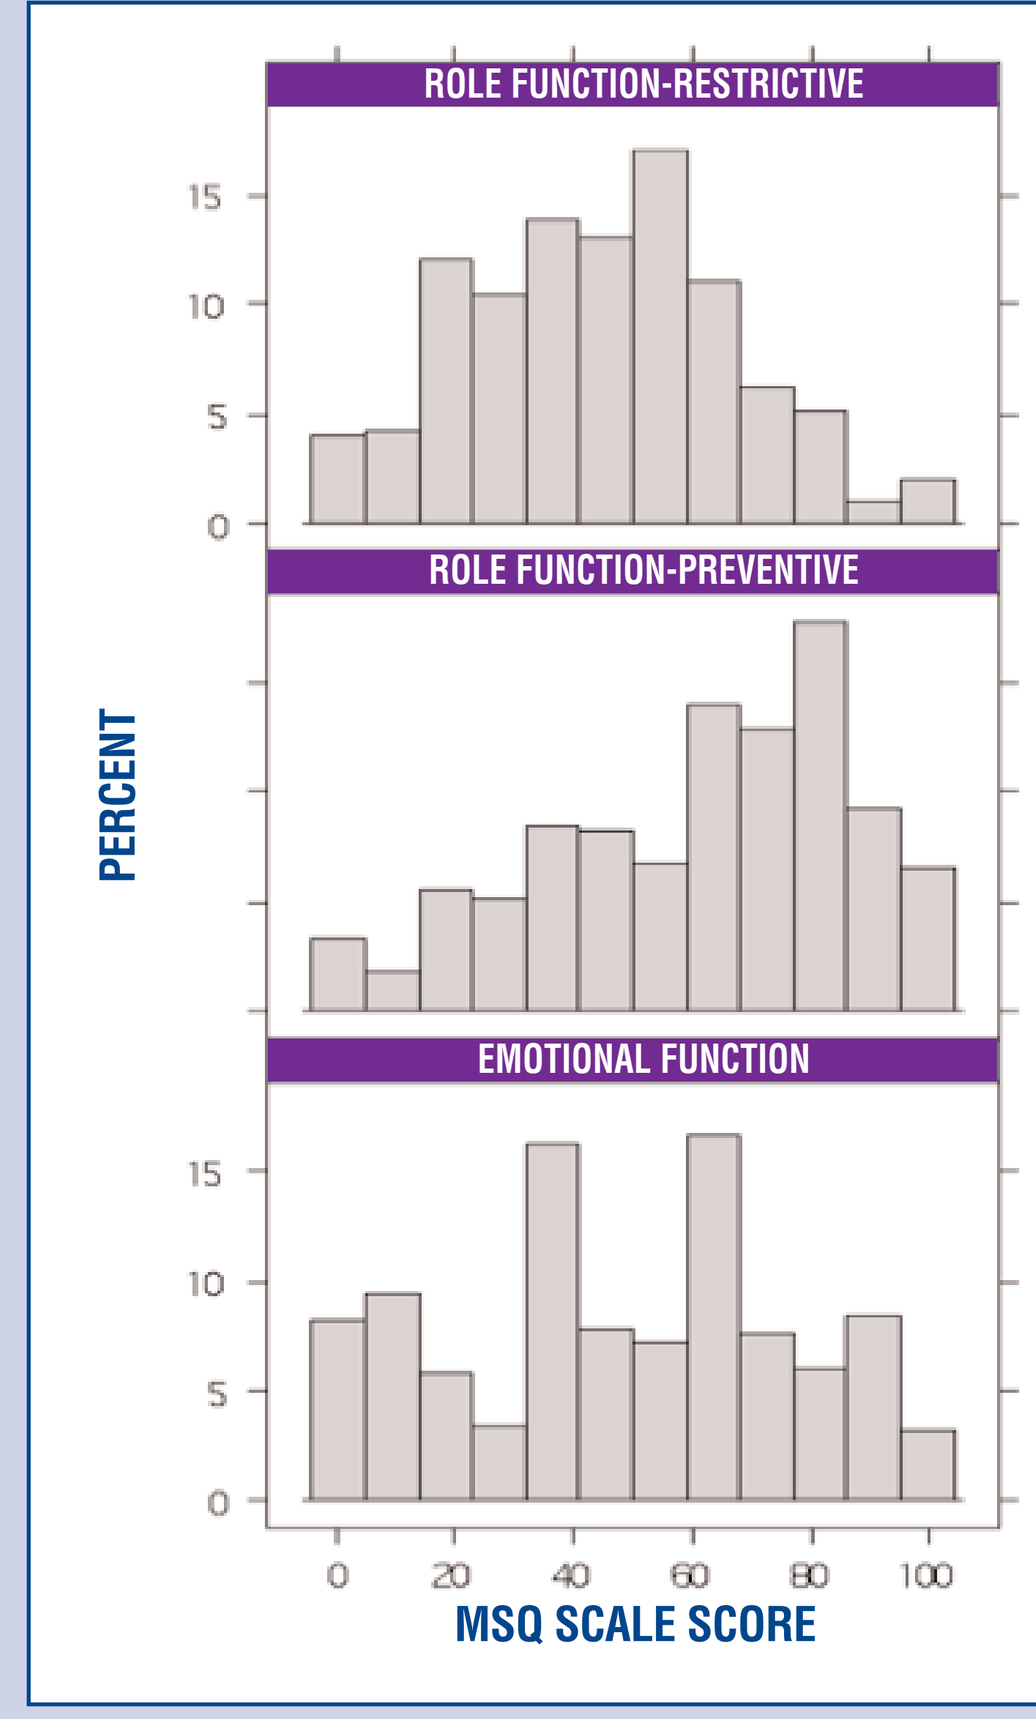
ROLE FUNCTION-RESTRICTIVE
ROLE FUNCTION-PREVENTIVE
EMOTIONAL FUNCTION
PERCENT
MSQ SCALE SCORE

TABLE 2. FACTOR LOADINGS FROM ALTERNATIVE CONFIRMATORY FACTOR ANALYSES MODELS EVALUATED IN CHRONIC MIGRAINE PATIENTS (N=499)

MSQ Item	Original 3-Factor Mode	1-Factor Model	MSQ 12 on RR Latent Factor
ROLE FUNCTION-RESTRICTIVE (RR)			
1 […] have migraines **interfered** with how well you dealt with family, friends and others who are close to you?	0.81	0.81	0.82
2 […] have migraines **interfered** with your leisure time activities, such as reading or exercising?	0.86	0.85	0.86
3 […] have you had **difficulty** in performing work or daily activities because of migraine symptoms?	0.94	0.92	0.93
4 […] did migraines **keep you** from getting as much done at work or at home?	0.89	0.88	0.89
5 […] did migraines **limit** your ability to concentrate on work or daily activities?	0.89	0.87	0.89
6 […] have migraines **left you too tired** to do work or daily activities?	0.80	0.80	0.80
7 […] have migraines **limited** the number of days you have felt energetic?	0.76	0.76	0.76
12 […] have you **felt** fed up or frustrated because of your migraines?	---	---	0.67
ROLE FUNCTION-PREVENTIVE (RP)			
8 […] have you had to **cancel** work or daily activities because you had a migraine?	0.85	0.78	0.86
9 […] did you **need help** in handling routine tasks such as every day household chores, doing necessary business, shopping, or caring for others, when you had a migraine?	0.79	0.73	0.79
10 […] did you have to **stop** work or daily activities to deal with migraine symptoms?	0.85	0.78	0.85
11 […] were you **not able to go** to social activities such as parties, dinner with friends, because you had a migraine?	0.85	0.77	0.85
EMOTIONAL FUNCTION (EF)			
12 […] have you **felt** fed up or frustrated because of your migraines?	0.72	0.68	---
13 […] have you **felt** like you were a burden on others because of your migraines?	0.87	0.68	0.89
14 […] have you been **afraid** of letting others down because of your migraines?	0.84	0.66	0.85
Chi-Square (DF)	229.22 (74)	568.25 (77)	239.78 (74)
CFI/TLI	0.97/0.96	0.89/0.87	0.96/0.96
RMSEA	0.07	0.11	0.07

TABLE 3. ITEM-SCALE CORRELATIONS CORRECTED FOR OVERLAP AND RELIABILITY STATISTICS FOR ORIGINAL THREE-FACTOR MODEL

	RR	RP	EF	Cronbach's Alpha	Cronbach's Alpha-Item deleted
ROLE FUNCTION-RESTRICTIVE (RR)				0.95	
1 […] have migraines **interfered** with how well you dealt with family, friends and others who are close to you?	0.79	0.67	0.64		0.94
2 […] have migraines **interfered** with your leisure time activities, such as reading or exercising?	0.84	0.70	0.59		0.94
3 […] have you had **difficulty** in performing work or daily activities because of migraine symptoms?	0.90	0.78	0.66		0.93
4 […] did migraines **keep you** from getting as much done at work or at home?	0.86	0.74	0.64		0.94
5 […] did migraines **limit** your ability to concentrate on work or daily activities?	0.86	0.70	0.64		0.94
6 […] have migraines **left you too tired** to do work or daily activities?	0.77	0.71	0.61		0.94
7 […] have migraines **limited** the number of days you have felt energetic?	0.75	0.61	0.62		0.95
ROLE FUNCTION-PREVENTIVE (RP)				0.90	
8 […] have you had to **cancel** work or daily activities because you had a migraine?	0.72	0.80	0.61		0.87
9 […] did you **need help** in handling routine tasks such as every day household chores, , doing necessary businessshopping, or caring for others, when you had a migraine?	0.68	0.74	0.57		0.89
10 […] did you have to **stop** work or daily activities to deal with migraine symptoms?	0.72	0.79	0.59		0.87
11 […] were you **not able to go** to social activities such as parties, dinner with friends, because you had a migraine?	0.71	0.80	0.62		0.87
EMOTIONAL FUNCTION (EF)				0.85	
12 […] have you **felt** fed up or frustrated because of your migraines?	0.67	0.55	0.63		0.86
13 […] have you **felt** like you were a burden on others because of your migraines?	0.62	0.64	0.77		0.73
14 […] have you been **afraid** of letting others down because of your migraines?	0.62	0.60	0.75		0.75

Note: Highlighted cells are hypothesized to be highest within the corresponding row

TABLE 4. TESTS OF DIFFERENTIAL ITEM FUNCTIONING ACROSS COUNTRY GROUPS (N=491; AUSTRALIA [N=55], CANADA [N=55], GERMANY [N=52], SPAIN [N=56], FRANCE [N=57], GREAT BRITAIN [N=57], ITALY [N=55], UNITED STATES [N=104])

	Tests of Non-Uniform DIF			Tests of Uniform DIF
Item	Chi-square	DF	P-value(1)	Percentage Change in Trait Level Coefficient(2)
ROLE FUNCTION-RESTRICTIVE				
1 […] have migraines **interfered** with how well you dealt with family, friends and others who are close to you?	5.499	7	0.5993	3.1
2 […] have migraines **interfered** with your leisure time activities, such as reading or exercising?	6.677	7	0.4632	0.4
3 […] have you had **difficulty** in performing work or daily activities because of migraine symptoms?	6.731	7	0.4574	1.4
4 […] did migraines **keep you** from getting as much done at work or at home?	3.366	7	0.8492	3.2
5 […] did migraines **limit** your ability to concentrate on work or daily activities?	15.715	7	0.0278	1.4
6 […] have migraines **left you too tired** to do work or daily activities?	4.256	7	0.7498	1.3
7 […] have migraines **limited** the number of days you have felt energetic?	12.428	7	0.0873	0.4
ROLE FUNCTION-PREVENTIVE				
8 […] have you had to **cancel** work or daily activities because you had a migraine?	6.130	7	0.5247	2.3
9 […] did you **need help** in handling routine tasks such as every day household chores, doing necessary business, shopping, or caring for others, when you had a migraine?	5.763	7	0.5677	7.4
10 […] did you have to **stop** work or daily activities to deal with migraine symptoms?	1.534	7	0.9811	1.1
11 […] were you **not able to go** to social activities such as parties, dinner with friends, because you had a migraine?	4.119	7	0.7659	4.6
EMOTIONAL FUNCTION				
12 […] have you **felt** fed up or frustrated because of your migraines?	14.958	7	0.0365	0.9
13 […] have you **felt** like you were a burden on others because of your migraines?	9.885	7	0.1952	1.1
14 […] have you been **afraid** of letting others down because of your migraines?	5.714	7	0.5735	3.3

(1) Non-uniform DIF was deemed to be present if the statistical significance of the interaction term between group membership and trait level was ≤ 0.05.
(2) Uniform DIF was deemed to be present if the change in the trait level coefficient resulting from removal of the group membership term from the model was >10%

TABLE 5. TESTS OF DIFFERENTIAL ITEM FUNCTIONING ACROSS ENGLISH/NON-ENGLISH SPEAKING COUNTRIES (N=499)

	Tests of Non-Uniform DIF			Tests of Uniform DIF
Item	Chi-square	DF	P-value(1)	Percentage Change in Trait Level Coefficient(2)
ROLE FUNCTION-RESTRICTIVE				
1 […] have migraines **interfered** with how well you dealt with family, friends and others who are close to you?	2.452	1	0.1174	0.3
2 […] have migraines **interfered** with your leisure time activities, such as reading or exercising?	1.434	1	0.2311	0.4
3 […] have you had **difficulty** in performing work or daily activities because of migraine symptoms?	1.193	1	0.2747	0.4
4 […] did migraines **keep you** from getting as much done at work or at home?	0.841	1	0.3592	2.6
5 […] did migraines **limit** your ability to concentrate on work or daily activities?	0.190	1	0.6626	0.0
6 […] have migraines **left you too tired** to do work or daily activities?	0.334	1	0.5635	0.1
7 […] have migraines **limited** the number of days you have felt energetic?	2.692	1	0.1009	0.1
ROLE FUNCTION-PREVENTIVE				
8 […] have you had to cancel work or daily activities because you had a migraine?	0.733	1	0.3919	0.0
9 […] did you **need help** in handling routine tasks such as every day household chores, doing necessary business, shopping, or caring for others, when you had a migraine?	0.055	1	0.8147	2.5
10 […] did you have to **stop** work or daily activities to deal with migraine symptoms?	0.031	1	0.8599	0.0
11 […] were you **not able to go** to social activities such as parties, dinner with friends, because you had a migraine?	0.503	1	0.4783	0.2
EMOTIONAL FUNCTION				
12 […] have you **felt** fed up or frustrated because of your migraines?	6.652	1	0.0099	0.1
13 […] have you **felt** like you were a burden on others because of your migraines?	0.502	1	0.4788	0.0
14 […] have you been **afraid** of letting others down because of your migraines?	2.762	1	0.0966	2.2

(1) Non-uniform DIF was deemed to be present if the statistical significance of the interaction term between group membership and trait level was ≤ 0.05.
(2) Uniform DIF was deemed to be present if the change in the trait level coefficient resulting from removal of the group membership term from the model was >10%

LIMITATIONS

- Electronic data collection may introduce participation bias because individuals lacking appropriate computer skills, access, or willingness may not have been included.
- Migraine diagnosis was not based on physician report but used the participants' self-report to the migraine screening questions. It is possible that some sufferers were misclassified.

CONCLUSIONS

- The 3-factor model currently proposed for the MSQ demonstrated appropriate psychometric characteristics among CM patients. The final model with three latent factors was found to have good model fit on all indices. Factor loadings were all greater than 0.70 and generally over 0.80. Item-level analyses indicated good psychometric properties for all, but one of the 14 items.
- Item 12 was found to correlate more strongly with the RR factor than with the hypothesized EF factor and was also found to be potentially associated with some degree of bias. Our findings regarding potential problems with item 12 agree with a prior study examining the validity of the MSQ in CM patients (7). Although the findings regarding the country bias of item 12 may suggest translation problems, removing item 12 from the EF scale did not improve model fit or increase the reliability of that scale.
- Among adults with chronic migraine, the MSQ provides a valid measure of RP, RR, and EF, yielding domain scores that can be reliably compared across languages and countries.

REFERENCES

(1) Lipton RB. Tracing transformation: chronic migraine classification, progression, and epidemiology. Neurology 2009; 72(5 Suppl):S3-S7.
(2) Bigal ME, Serrano D, Reed M, Lipton RB. Chronic migraine in the population: burden, diagnosis, and satisfaction with treatment. Neurology 2008; 71(8):559-566.
(3) Jhingran P, Osterhaus JT, Miller DW, Lee JT, Kirchdoerfer L. Development and validation of the Migraine-Specific Quality of Life Questionnaire 151. Headache 1998; 38(4):295-302.
(4) Martin BC, Pathak DS, Sharfman MI, Adelman JU, Taylor F, Kwong WJ et al. Validity and reliability of the Migraine-Specific Quality of Life Questionnaire (MSQ Version 2.1). Headache 2000; 40(3):204-15.
(5) Jhingran P, Davis SM, LaVange LM, Miller DW, Helms RW. MSQ: Migraine-Specific Quality-of-Life Questionnaire. Further investigation of the factor structure 152. Pharmacoeconomics 1998; 13(6):707-717.
(6) Crane PK, Gibbons LE, Jolley L, van Belle G. Differential item functioning analysis with ordinal logistic regression techniques. DIFdetect and difwithpar. Med Care 2006;44:S115-S123.
(7) Cole JC, Lin P, Rupnow MF. Validation of the Migraine-Specific Quality of Life Questionnaire version 2.1 (MSQ v. 2.1) for patients undergoing prophylactic migraine treatment. Qual Life Res 2007; 16(7):1231-1237.

DISCLOSURES

This research was sponsored by Allergan, Inc., Irvine, CA.

SFV and GAM are employees of Allergan, Inc.

Poster presented at: the ISPOR 15th Annual International Meeting; May 15-19, 2010; Atlanta, GA.